EHA2025 Congress
June 12-15 | Milan, Italy

The phase I/II TG01-study: Vaccinating against RAS-mutated Multiple Myeloma

Hanne Marie Norseth MD[1,2], Hedda Berg Monsen[1,2], Nora Remen[1], Else Marit Inderberg[3], Helen Vålerhaugen[4], Laila Helene Bergly[5], Emilie Einertsen[3], Anna Lysén PhD[1], Ingerid Weum Abrahamsen MD, PhD[1], Fredrik Schjesvold, MD, PhD[1]

[1]Oslo Myeloma Center, Department of Hematology, Oslo University Hospital, Oslo, Norway; [2]Institute of Clinical Medicine, Faculty of Medicine, University of Oslo, Norway; [3]Translational Research Unit, Department of Cellular Therapy, Oslo University Hospital, Norway; [4]Department of Pathology, Oslo University Hospital, Norway, [5] Section for Cancer Cytogenetics, Institute for Cancer Genetics and Informatics, Oslo University Hospital, Oslo, Norway

UNIVERSITY OF OSLO

INTRODUCTION

- RAS mutations are key genomic driver events in the development and progression of multiple myeloma (MM) (1).
- TG01 is an injectable mixture of 7 different synthetic peptides that represent fragments of mutant forms of the human RAS proteins (2).
- The study (NCT05841550) is evaluating vaccination with TG01, given with QS-21 as an adjuvant, as single agent in patients with either MM with measurable disease following at least one previous line of therapy or in high-risk smoldering MM (HR SMM) whose tumor carry KRAS or NRAS codon 12/13 mutations.

AIMS

- **Primary endpoint:** safety and tolerability of TG01/QS-21
- **Secondary endpoints:** response according to IMWG criteria and immunological response to the vaccine defined by the TG01-specific cytokine production

METHODS

- 20 patients will be included and receive TG01/QS-21 0,7 mg SC Q2W for the first 12 weeks followed by Q2M until week 52 for a total of 12 doses. Patients are screened for RAS mutations and only included if they have one of the 7 mutations (12A, 12C, 12D, 12R, 12S, 12V, 13D) that are targeted by the TG01 vaccine.
- The immune response of PBMCs from patients' blood against the TG01 vaccine is evaluated using the Human IFN-y Single-Color ELISPOT assay.

RESULTS

- As of May 12, 2025, 63 patients have been screened and 12 patients have been included since the time of study initiation May 31, 2023, with reasons for screen failure being no relevant RAS mutation (n=47), newly diagnosed MM according to IMWG (=6) and no measurable disease (n=1).
- 6 patients (50%) have HR SMM and 6 patients (50%) have MM with a median of 2 (range 1-3) prior lines of therapy. The median age is 72.5 years (range 58-82) with 9/12 patients (75%) being male and 3/12 (25%) being female.
- **Safety:** 10 patients (83%) have experienced a treatment-emergent adverse event with flu-like symptoms (n=6; 50%), local skin reaction (n=2; 17%), chills (n=2, 17%) and covid-19 infection (n=2, 17%) being the most common. No patients have developed $\geq$ grade 3 AEs.
- **Clinical responses:** With a median FU of 19 months (range 4-23) 6/12 patients (50%) have discontinued the study due to disease progression. For the 6 patients still on study, all have stable disease (SD) with a median FU of 13 months (range 4-20 months). The median PFS in all 12 evaluable patients at data cutoff is not reached.
- **Immunological responses:** Variable frequencies of T-cells reacting against peptides corresponding to mutant variants of K/N-RAS were analyzed by ELISPOT assay in PBMCs of all 12 patients at baseline. During the course of vaccination, the frequencies increased ≥2-fold in 6/12 patients (Fig.1 and Fig.2), and these were qualified as immunological responders to TG01/QS21. The response in patient TG01-037 was just narrowly below the qualification cutoff (Fig.1 and Fig.2). 4 of the patients with vaccine-induced T-cell responses, along with patient TG01-037 (5 in total), are amongst the 6 patients remaining on study. All patients except one demonstrated higher post-vaccination responses against the TG01 vaccine than against the K/N-WT peptide at most of the time points (Fig.2 and Fig.3) Frequencies of K/N-RAS-specific T-cells at disease progression dropped below the respective baseline levels in 4/6 PD patients.

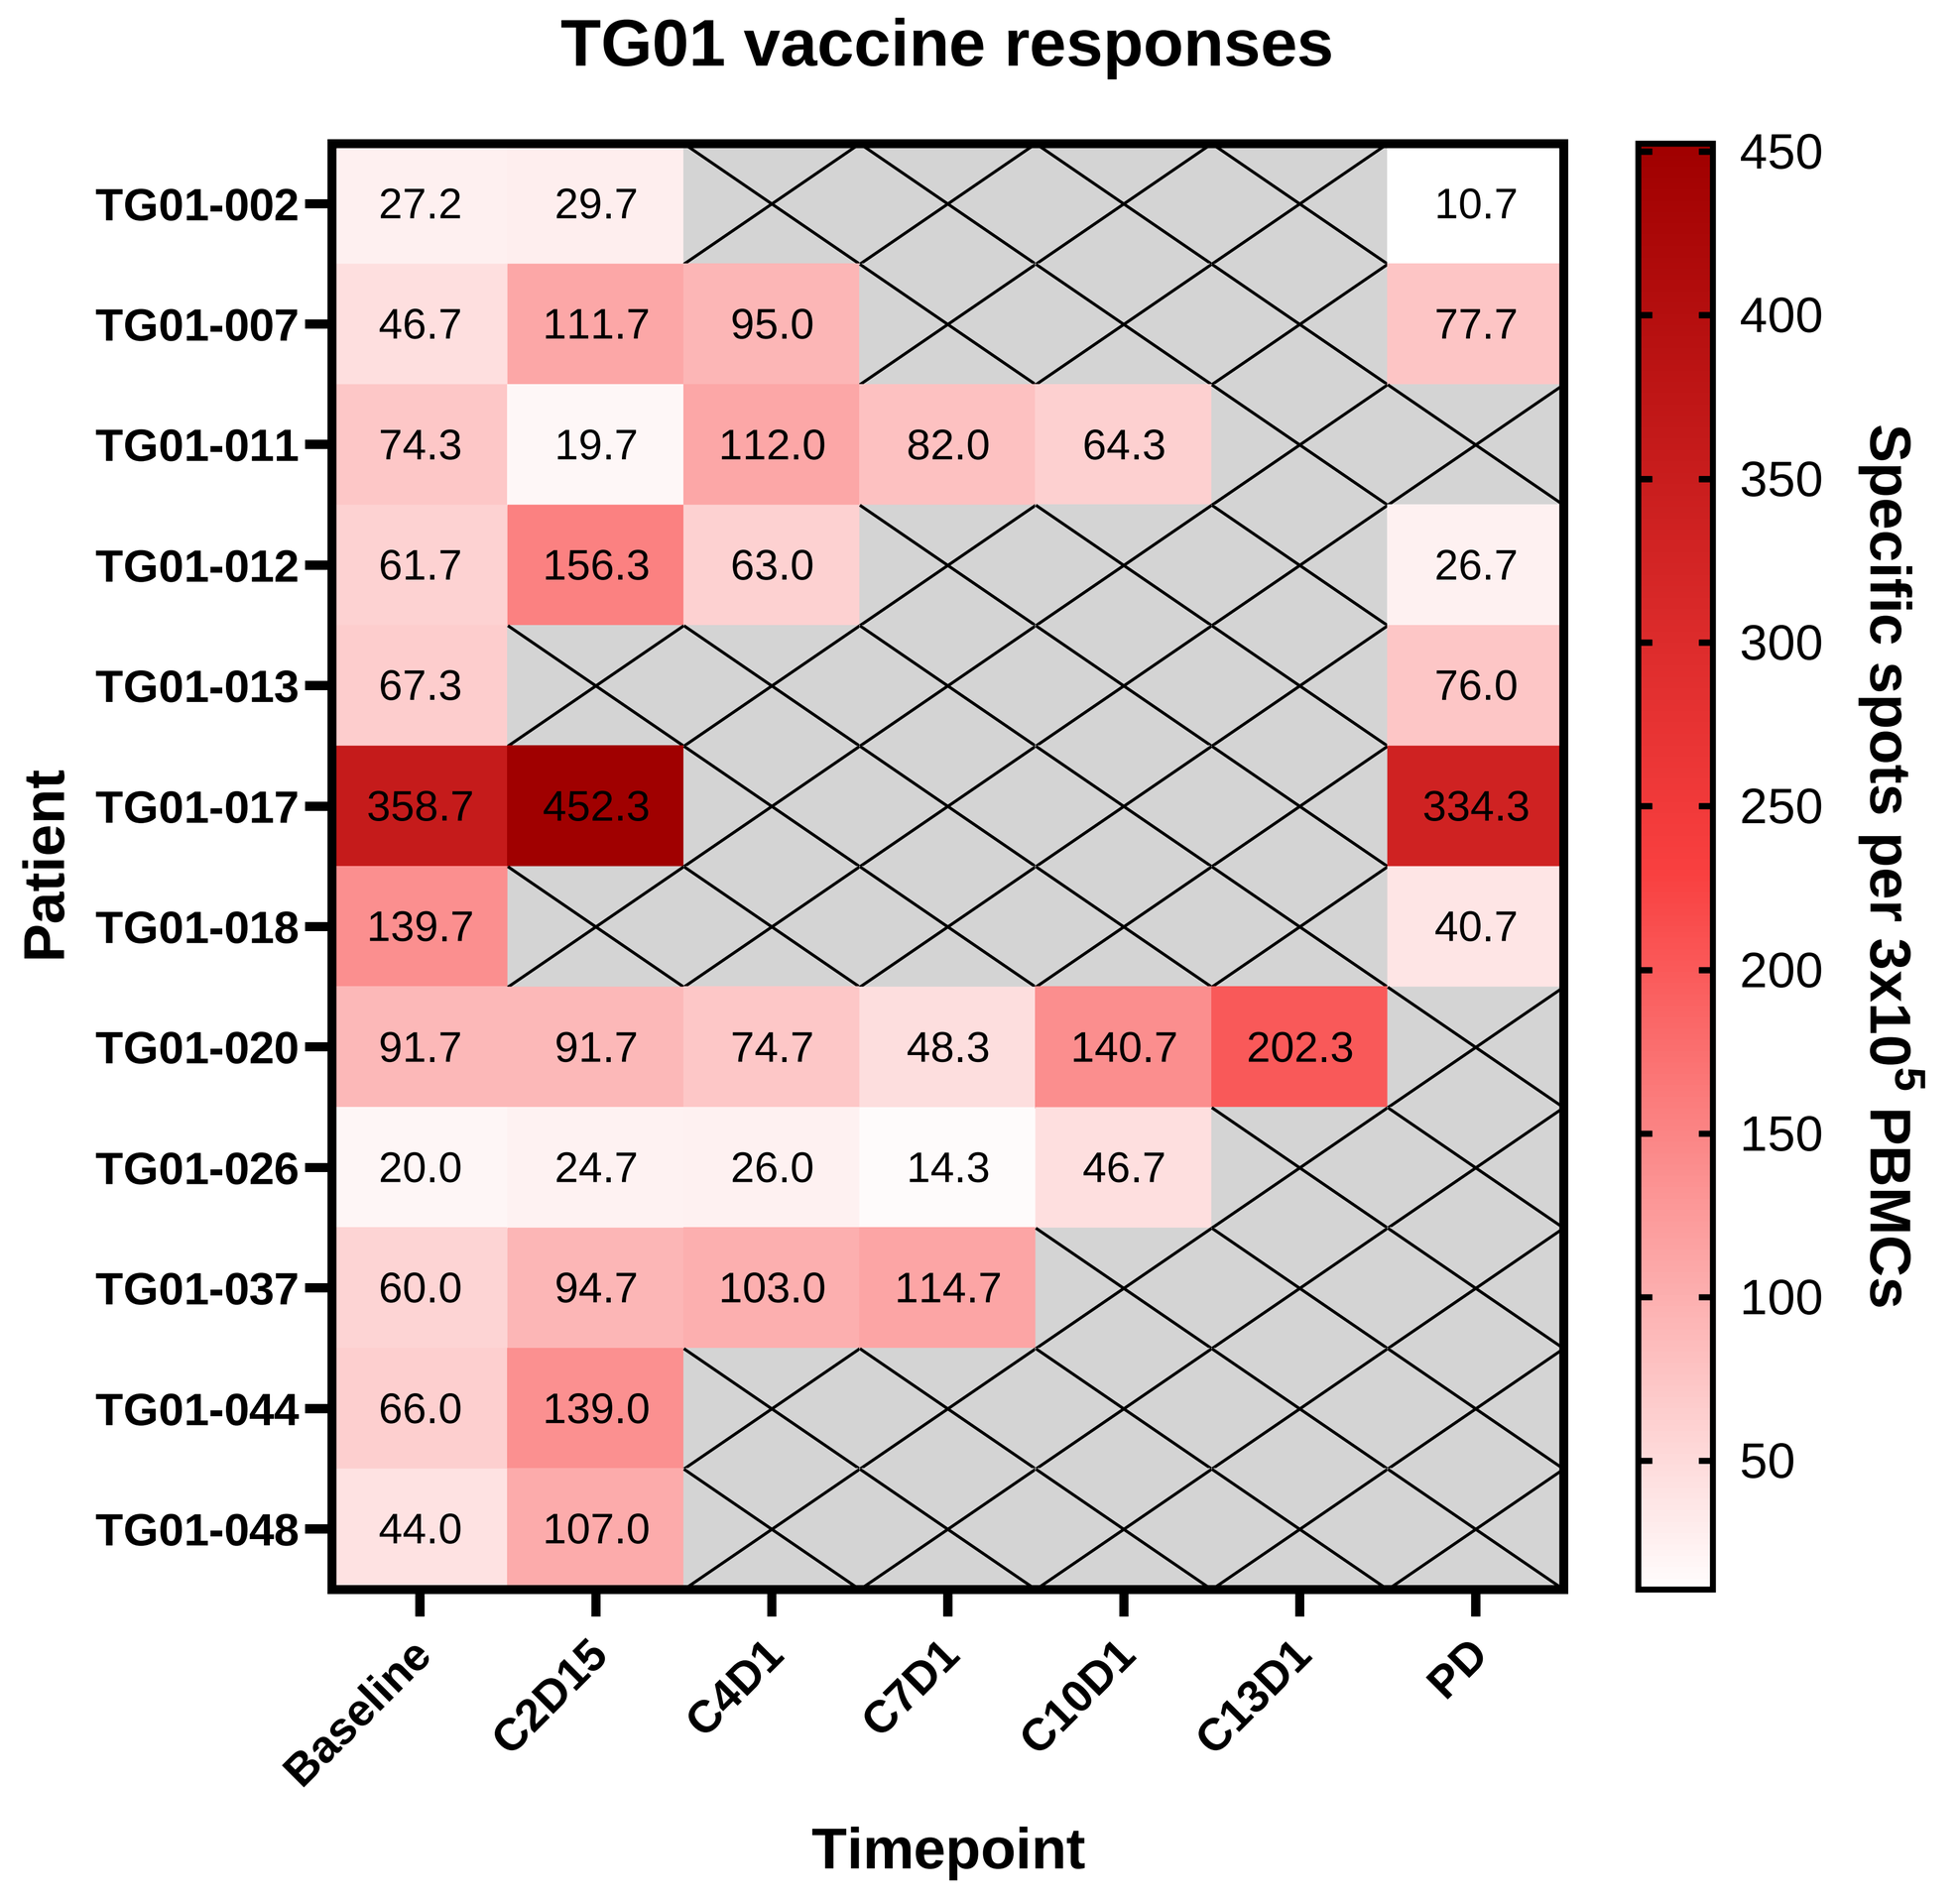
TG01 vaccine responses
Patient
TG01-002 27.2 29.7 10.7
TG01-007 46.7 111.7 95.0 77.7
TG01-011 74.3 19.7 112.0 82.0 64.3
TG01-012 61.7 156.3 63.0 26.7
TG01-013 67.3 76.0
TG01-017 358.7 452.3 334.3
TG01-018 139.7 40.7
TG01-020 91.7 91.7 74.7 48.3 140.7 202.3
TG01-026 20.0 24.7 26.0 14.3 46.7
TG01-037 60.0 94.7 103.0 114.7
TG01-044 66.0 139.0
TG01-048 44.0 107.0
Baseline
C2D15
C4D1
C7D1
C10D1
C13D1
PD
Timepoint
450
400
350
300
250
200
150
100
50
Specific spots per 3x10^5 PBMCs

Fig. 1 shows the frequency of IFN-g producing T cells in response to a 48-hour *ex vivo* stimulation of peripheral blood mononuclear cells (PBMC) with the TG01 vaccine peptides. The intensity of the heatmap is increased with increasing frequencies of spot counts in each sample. PBMCs from each time point were tested in triplicates and the average spot counts for each time point is shown, minus the background for PBMCs with no peptide stimulation.

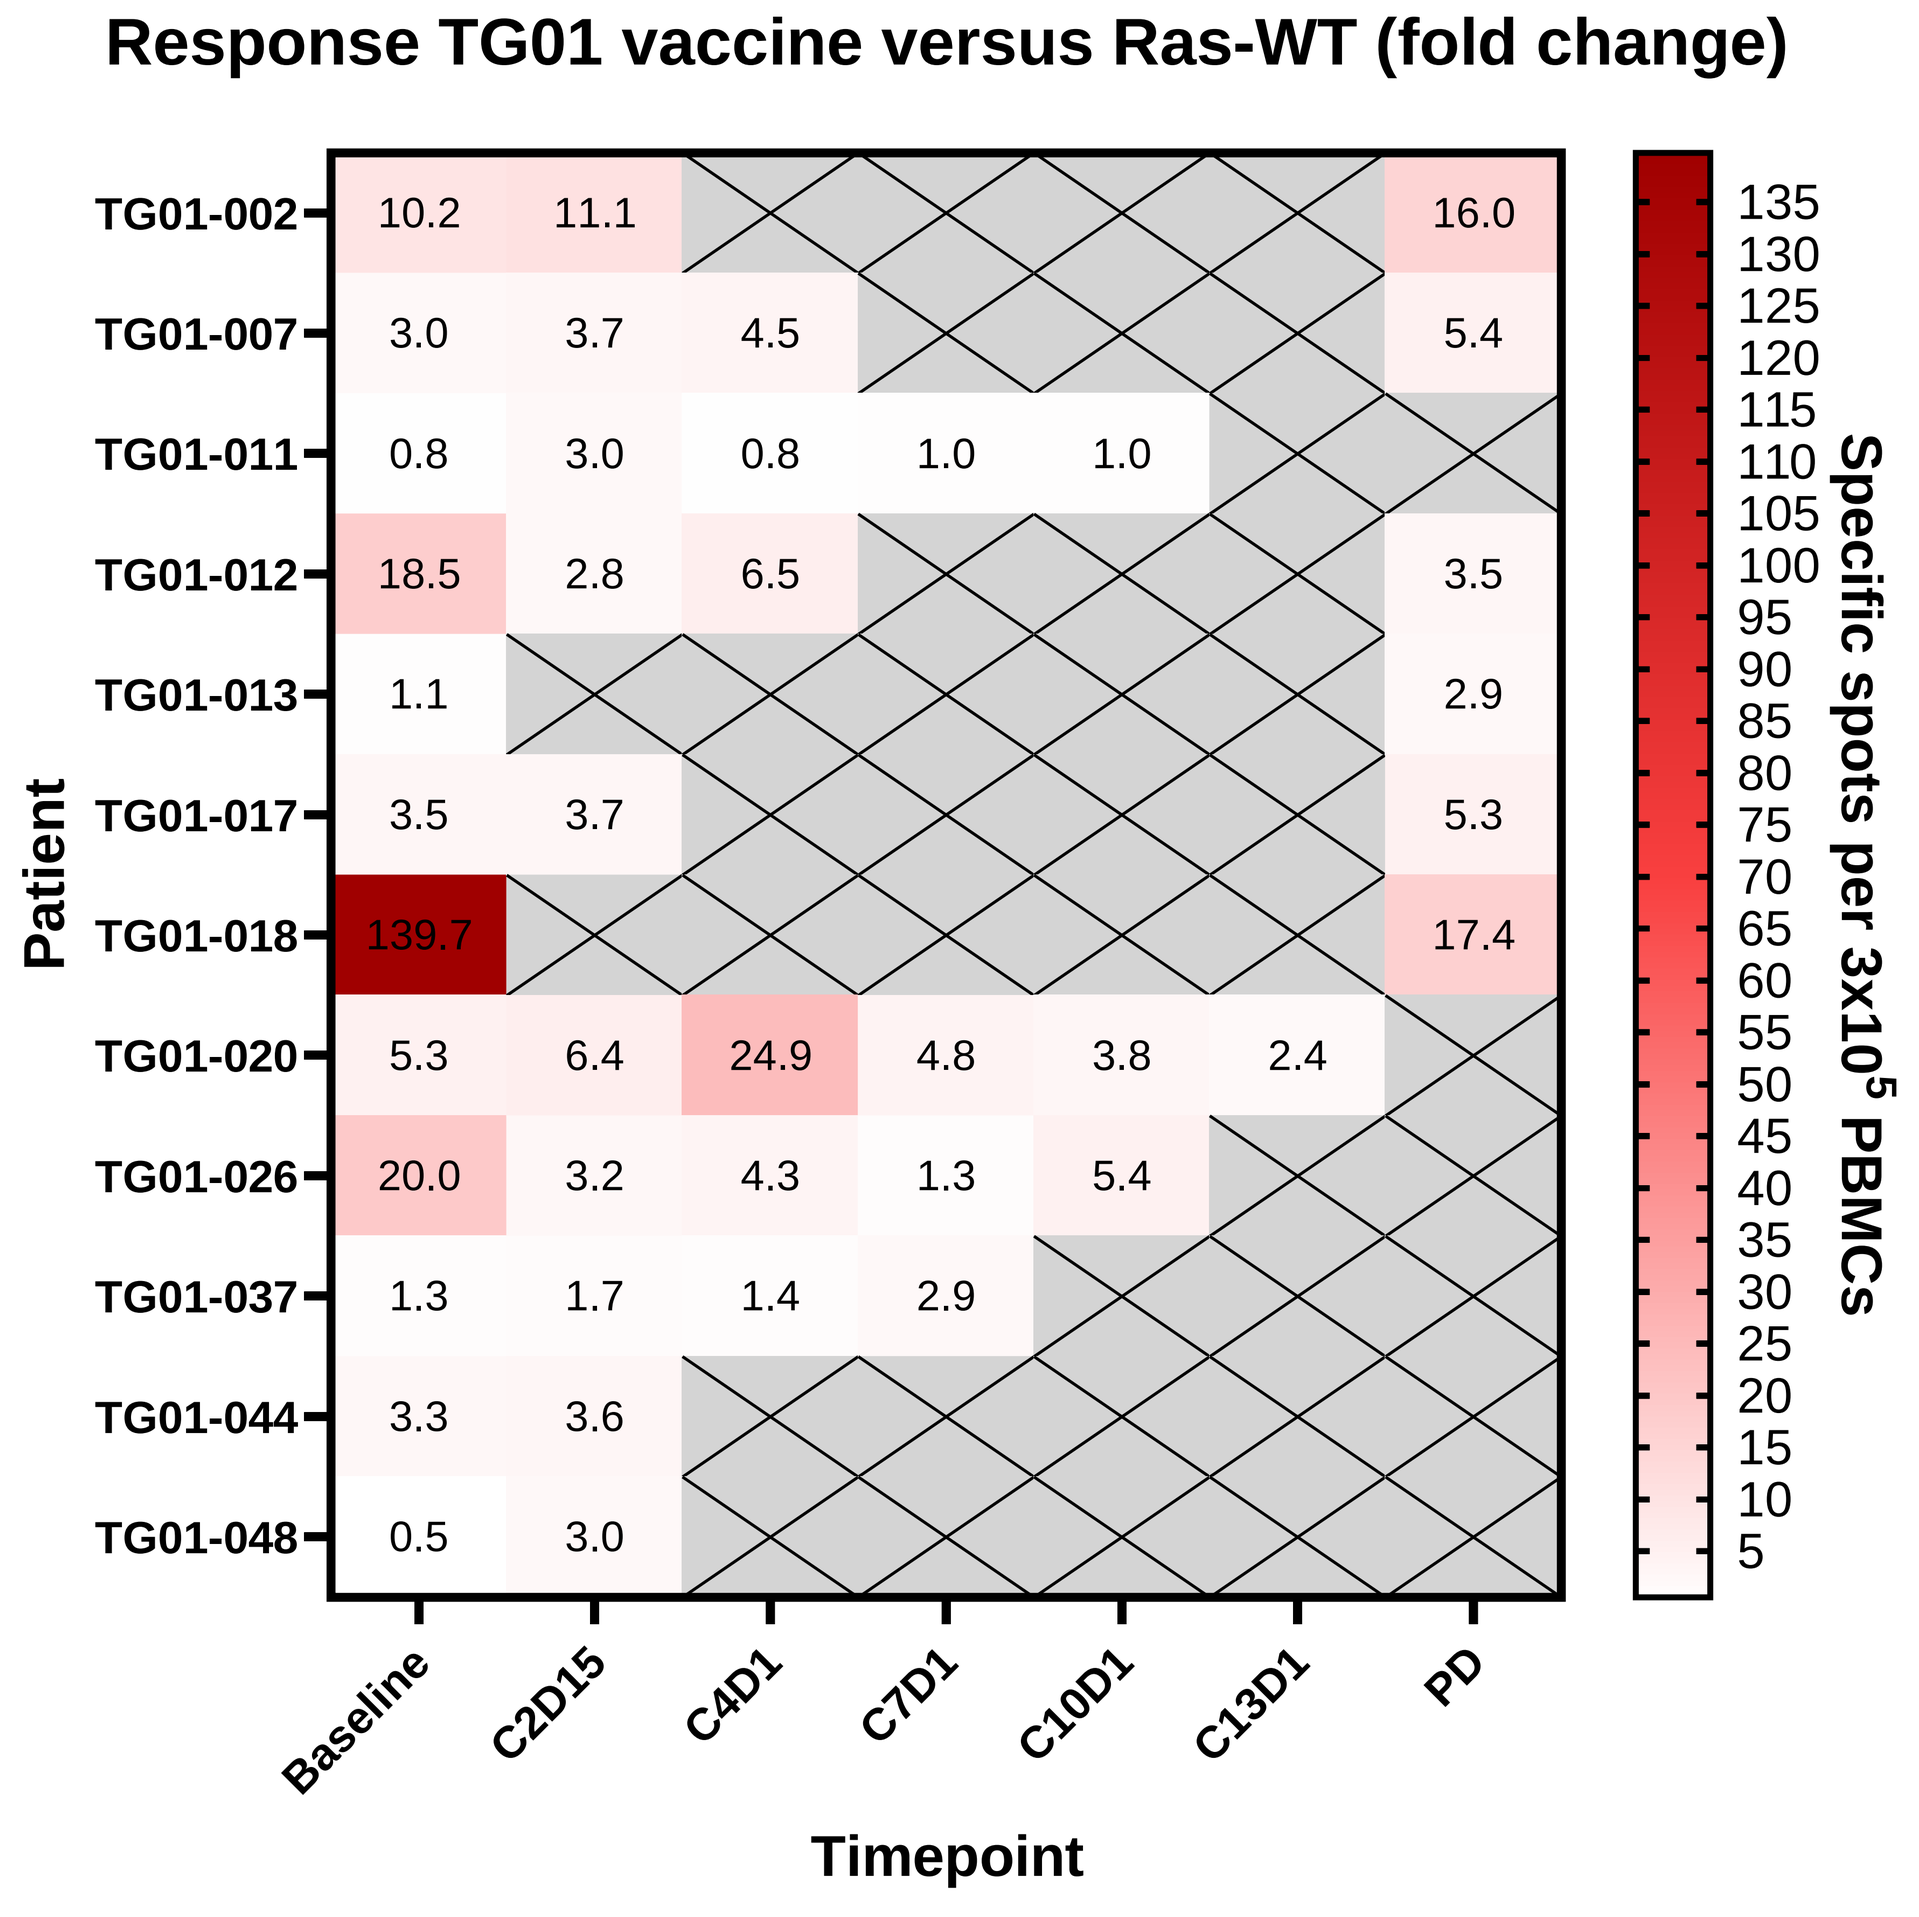
Response TG01 vaccine versus Ras-WT (fold change)
Patient
TG01-002 10.2 11.1 16.0
TG01-007 3.0 3.7 4.5 5.4
TG01-011 0.8 3.0 0.8 1.0 1.0
TG01-012 18.5 2.8 6.5 3.5
TG01-013 1.1 2.9
TG01-017 3.5 3.7 5.3
TG01-018 139.7 17.4
TG01-020 5.3 6.4 24.9 4.8 3.8 2.4
TG01-026 20.0 3.2 4.3 1.3 5.4
TG01-037 1.3 1.7 1.4 2.9
TG01-044 3.3 3.6
TG01-048 0.5 3.0
Baseline
C2D15
C4D1
C7D1
C10D1
C13D1
PD
Timepoint
135 130 125 120 115 110 105 100 95 90 85 80 75 70 65 60 55 50 45 40 35 30 25 20 15 10 5
Specific spots per 3x10^5 PBMCs

Fig. 2 shows the fold change in stimulation between PBMCs stimulated with the TG01 vaccine peptides versus the corresponding non-mutated Ras wild type (Ras-WT) peptide.

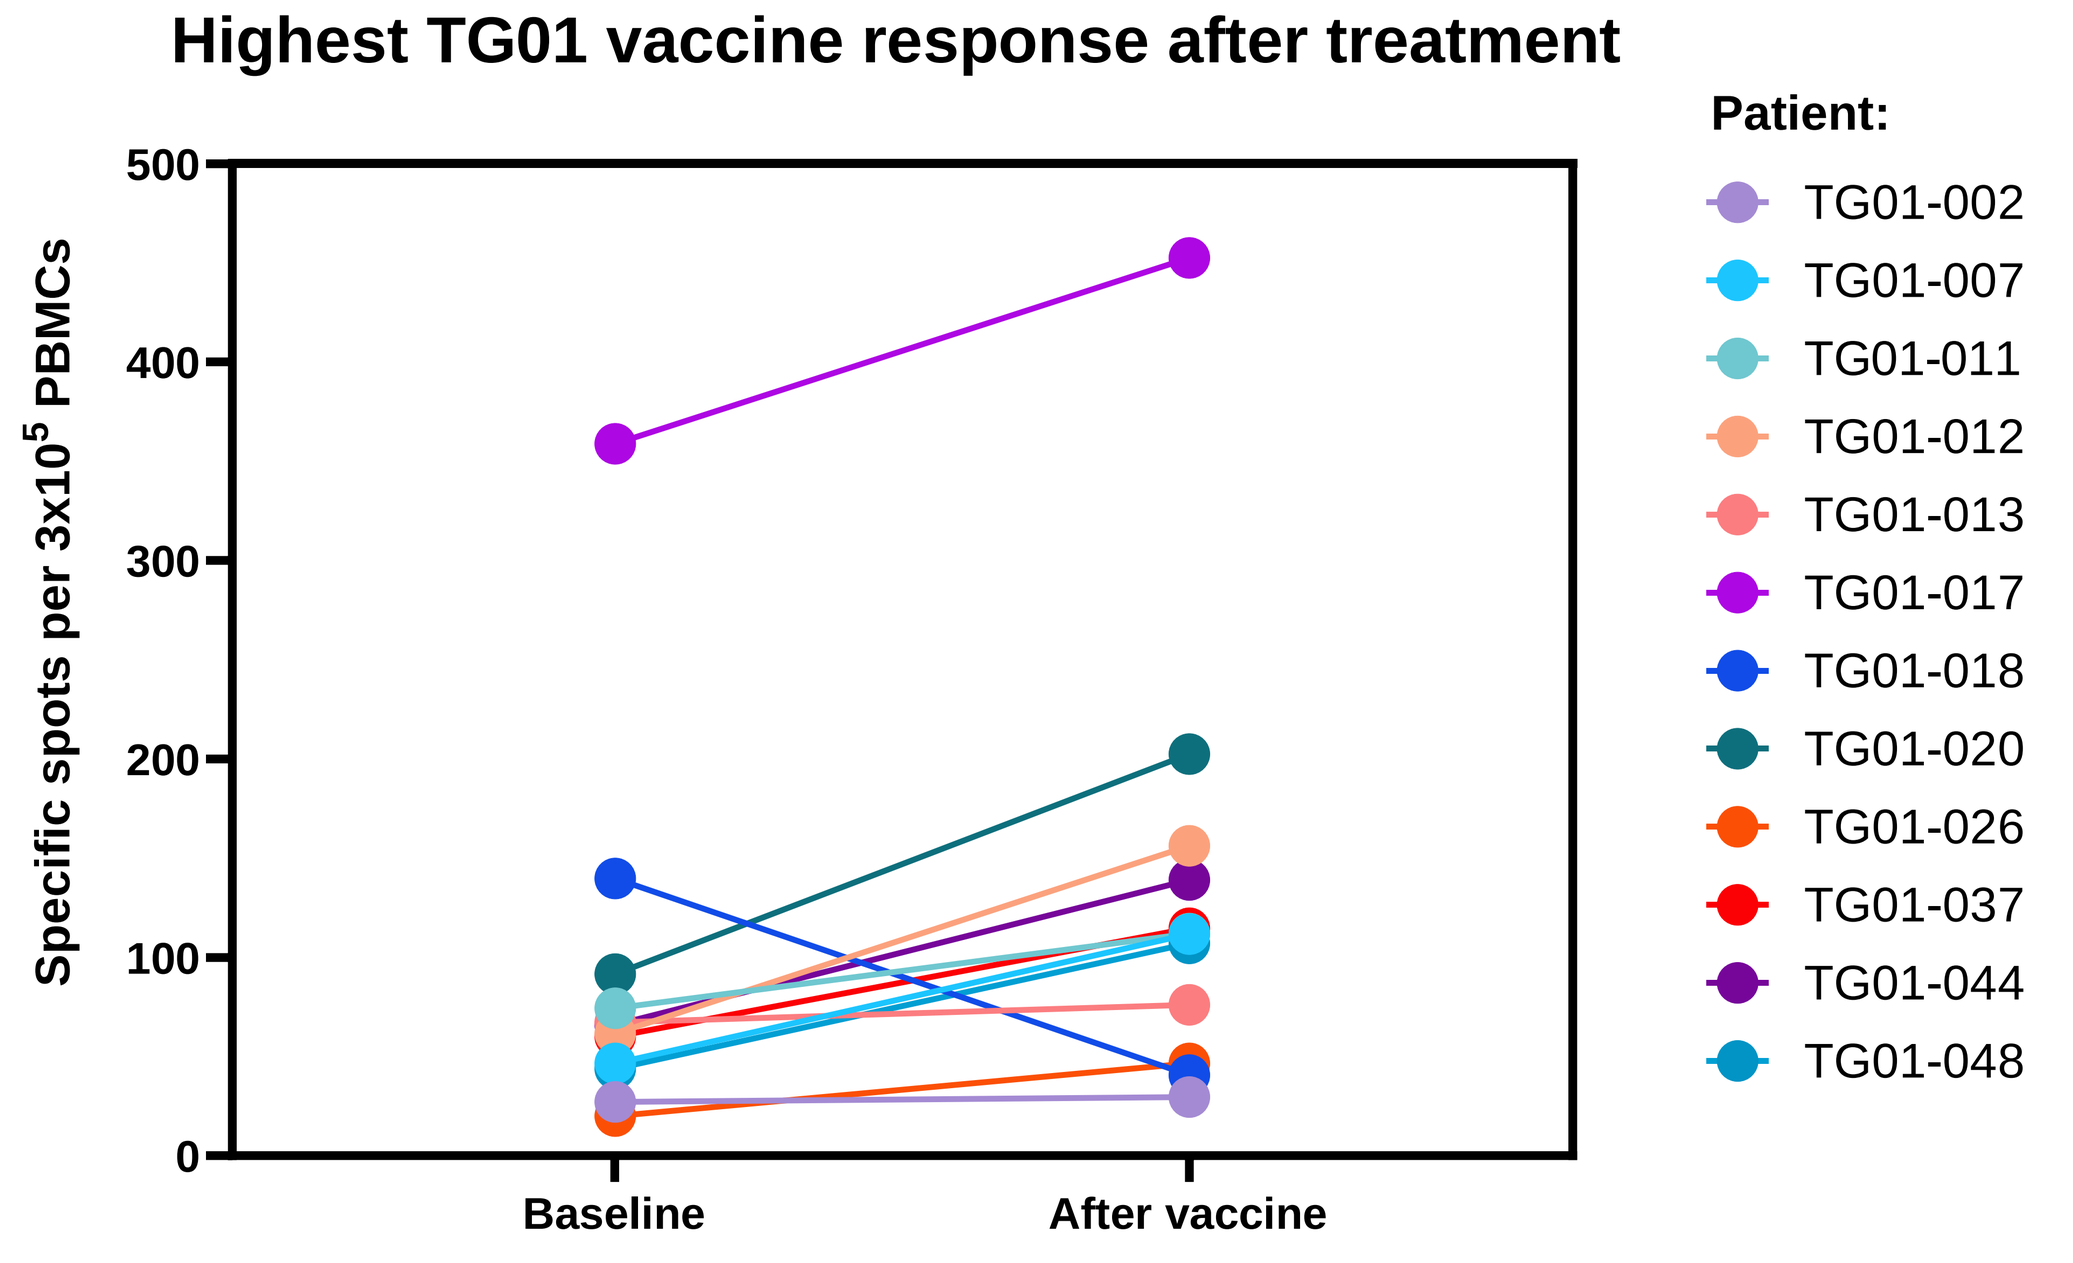
Highest TG01 vaccine response after treatment
Specific spots per 3x10^5 PBMCs
500
400
300
200
100
0
Baseline
After vaccine
Patient:
TG01-002
TG01-007
TG01-011
TG01-012
TG01-013
TG01-017
TG01-018
TG01-020
TG01-026
TG01-037
TG01-044
TG01-048

Fig. 3 shows the baseline response and the highest post-vaccination response measured in IFN-g ELISPOT (see figure 1).

An increased response post-vaccination was seen in all patients except for patient TG01-18 who had a high baseline respons, but then progressed very rapidly correlating with a reduction in the immune response.

CONCLUSIONS

- Available data demonstrate excellent tolerability and safety of TG01/QS-21 vaccination.
- 50% (6/12) of vaccinated patients show vaccine-induced specific T-cell responses against mutant K/N-RAS-peptides.
- 50% (6/12) of patients remain on study with SD, no objective responses have so far been observed.
- 67% (4/6) of patients with SD had a K/N-RAS-peptide specific immune response by ELISPOT (1/2 negative patients fell very narrowly below positivity threshold)
- Enrollment and analysis of the TG01 vaccine-specific responses are ongoing.

REFERENCES

(1) Boyle, E.M., Deshpande, S., Tytarenko, R. et al. The molecular make up of smoldering myeloma highlights the evolutionary pathways leading to multiple myeloma. Nat Commun 12, 293 (2021).

(2) Palmer, D.H et al. TG01/GM-GSF and adjuvant gemcitabine in patients with resected RAS-mutant adenocarcinoma of the pancreas (CT TG01-01): a single-arm, phase 1/2 trial. Br J Cancer 122, 971–977 (2020).

ACKNOWLEDGEMENT

We thank Circio Holding ASA for funding this research.

CONTACT INFORMATION

Hanne Marie Liddle Norseth

hanors@ous-hf.no

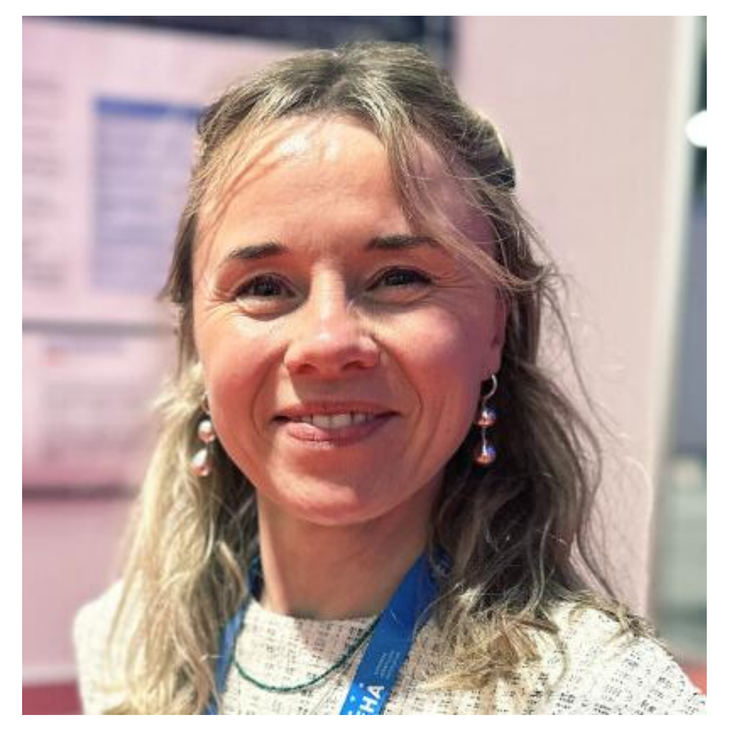